Integrated Editing and Manufacturing Process Design for BEAM-101, an Autologous CD34+ HSPC Therapy for Sickle Cell Disease, Results in Robust Process Yield and Increased HbF Induction

Corrina Lucini[1,2], Lillian Ye[1,2], Brent Coisman[1,2], John Jamieson[1], Margaret Zhang[1], Jordan Kim[1], Nate Goff[1], Brianna Mccuaig[1], Kathryn Goldner[1], Jack Mai[1], Jessica Hoyer[1], Alexandra Shay[1], Ajit Kamath[1], Kelly Fitzpatrick[1], Tao Bai[1], Kangjian Qiao[1,3], Christopher Oesterling[1,3], S. Haihua Chu[1,3], Paul Kopesky[1,3]

[1]Beam Therapeutics, Cambridge, MA, United States of America; [2]These authors contributed equally; [3]Co-senior authors

1. Introduction

- ❖The development of autologous cell therapies for sickle cell disease (SCD) is inherently complex and requires integration of both product and process design.
- ❖This integration includes not only the development of CD34+ cell isolation and editing manufacturing processes that have the potential to improve yield, reduce the number of mobilization cycles, and shorten the processing duration but also careful selection of gene editing methodology that maximizes the desired therapeutic outcomes.
- ❖Currently, re-expression of fetal hemoglobin (HbF) can be achieved through distinct mechanisms of action including base editing of HBG1/2, Cas9-nuclease editing of HBG1/2, or Cas9-nuclease disruption of BCL11A expression.
- ❖A comprehensive approach to therapeutic and process design that optimizes the induction of HbF and optimizes manufacturing process automation has the potential to improve product quality and the overall therapeutic experience for patients.

2. CD34+ HSCT Therapy for Sickle Cell Disease

- ❖ BEAM-101: Designed to be best-in-class genetic medicine for SCD

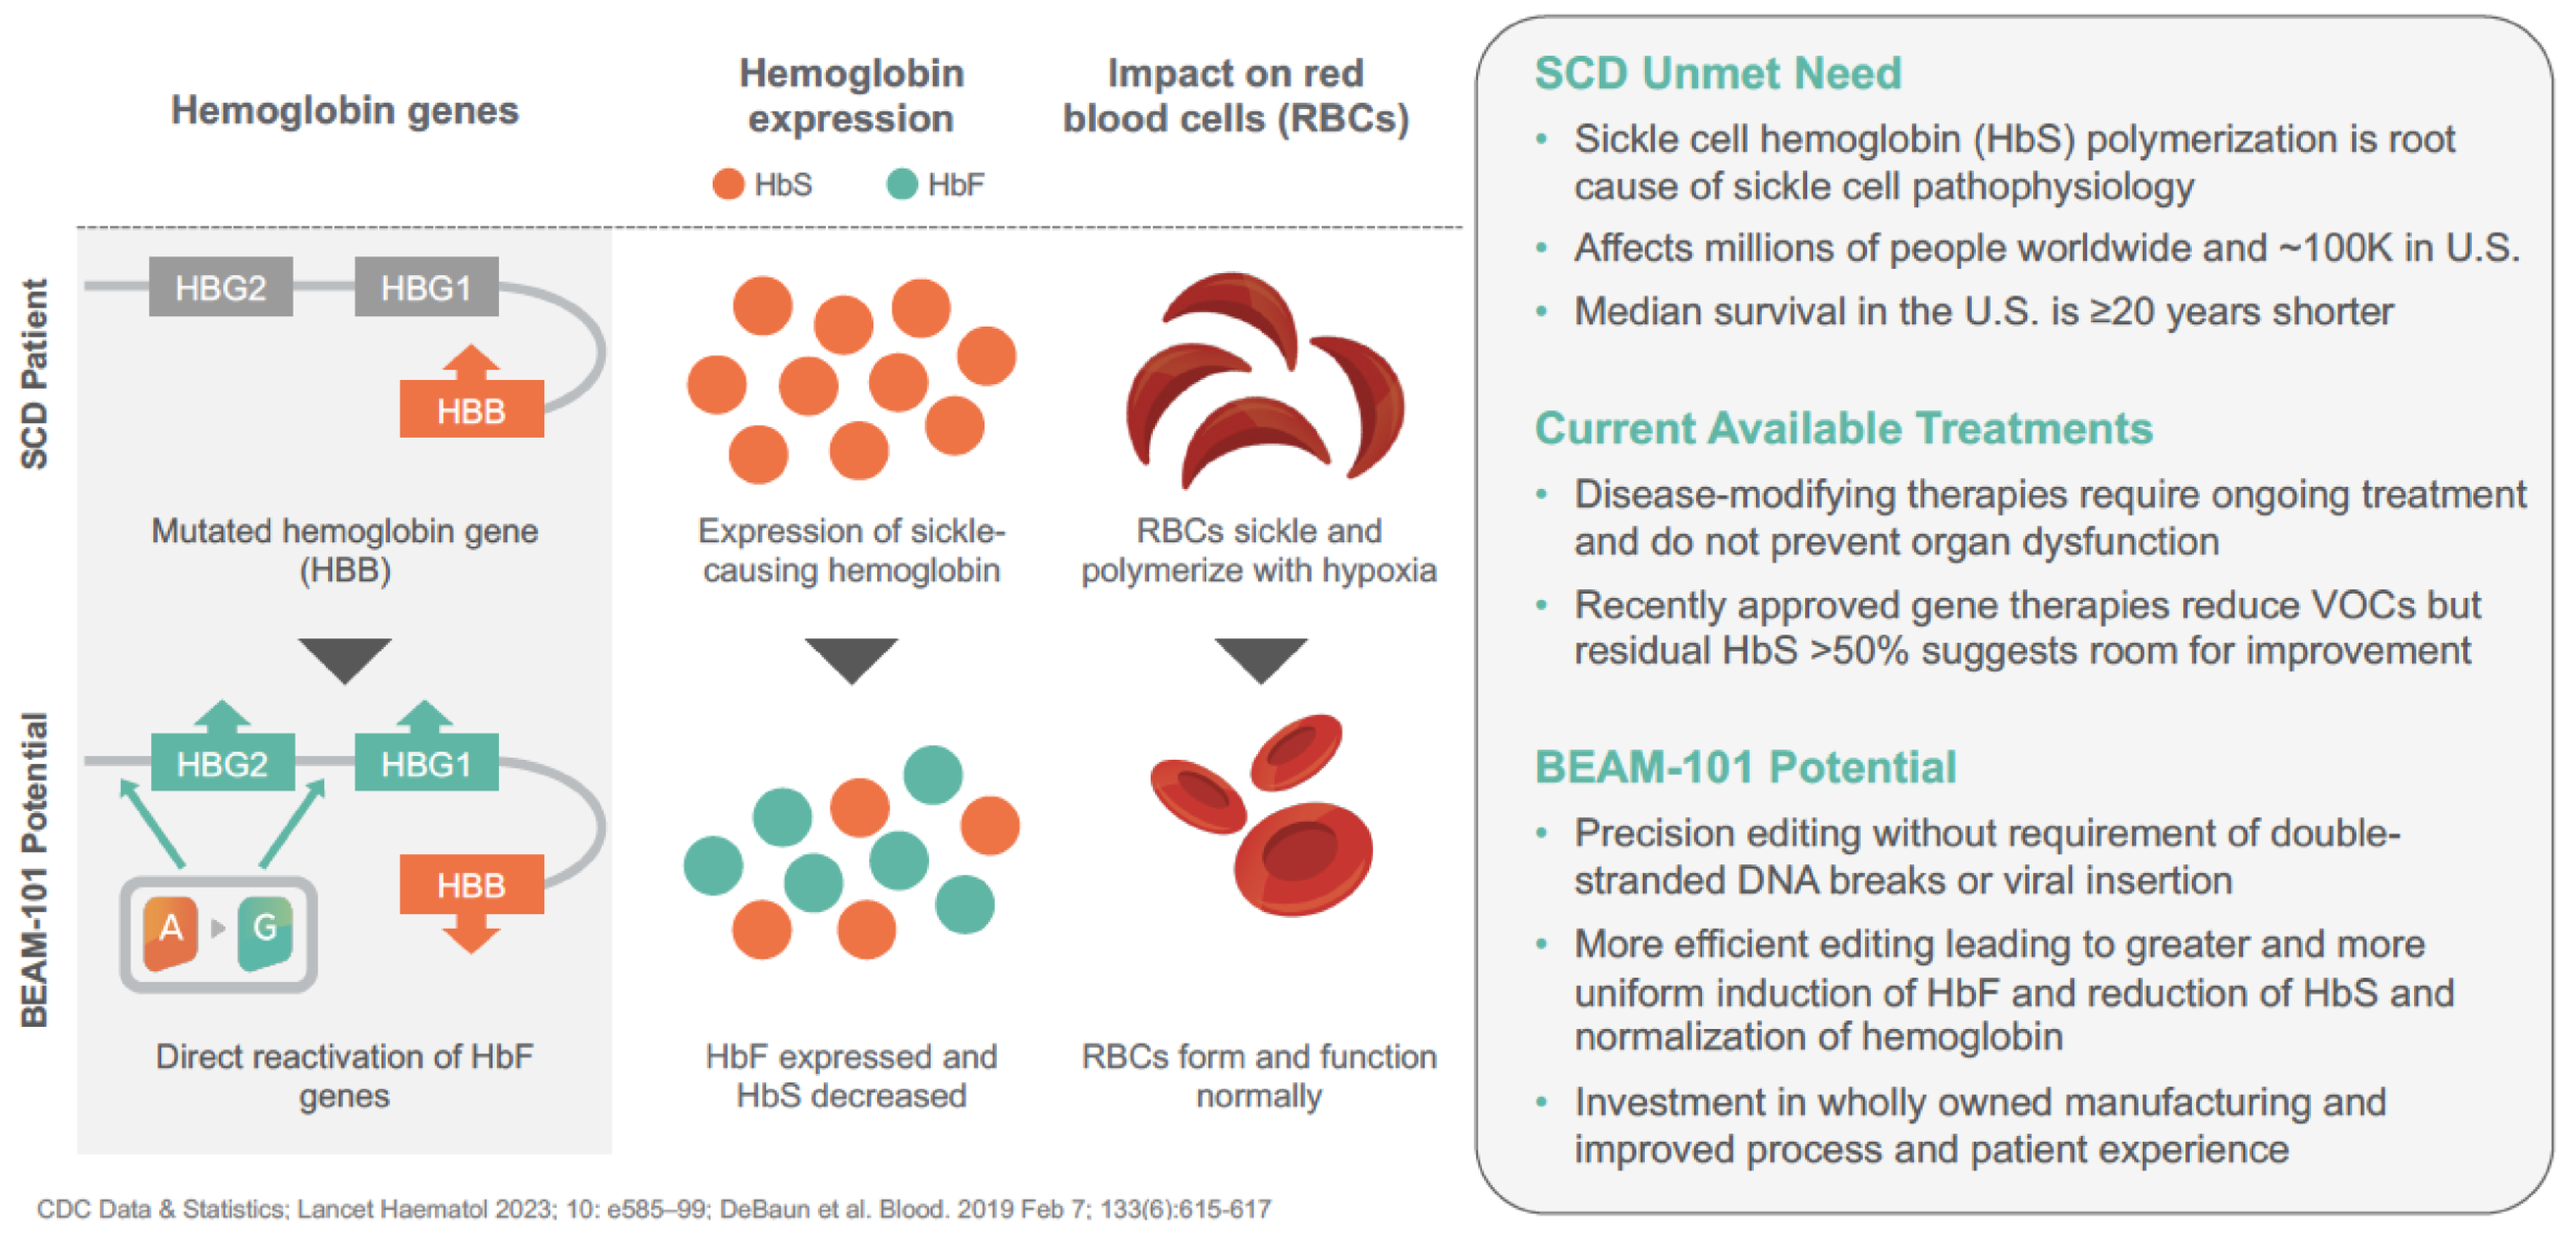
Hemoglobin genes
Hemoglobin expression
HbS
HbF
Impact on red blood cells (RBCs)
SCD Patient
HBG2
HBG1
HBB
Mutated hemoglobin gene (HBB)
Expression of sickle-causing hemoglobin
RBCs sickle and polymerize with hypoxia
BEAM-101 Potential
A
G
Direct reactivation of HbF genes
HbF expressed and HbS decreased
RBCs form and function normally
SCD Unmet Need
Sickle cell hemoglobin (HbS) polymerization is root cause of sickle cell pathophysiology
Affects millions of people worldwide and ~100K in U.S.
Median survival in the U.S. is ≥20 years shorter
Current Available Treatments
Disease-modifying therapies require ongoing treatment and do not prevent organ dysfunction
Recently approved gene therapies reduce VOCs but residual HbS >50% suggests room for improvement
BEAM-101 Potential
Precision editing without requirement of double-stranded DNA breaks or viral insertion
More efficient editing leading to greater and more uniform induction of HbF and reduction of HbS and normalization of hemoglobin
Investment in wholly owned manufacturing and improved process and patient experience

CDC Data & Statistics; Lancet Haematol 2023; 10: e585–99; DeBaun et al. Blood. 2019 Feb 7; 133(6):615-617

3. Objective

- ❖ The aims of this study were 1) to evaluate base-editing and nuclease-editing methodologies to both increase re-expression of HbF and minimize cell damage and repair pathway activation 2) to evaluate the capability of an automated process to manufacture high drug product quality for use in the BEACON Phase 1/2 clinical trial (NCT05456880).

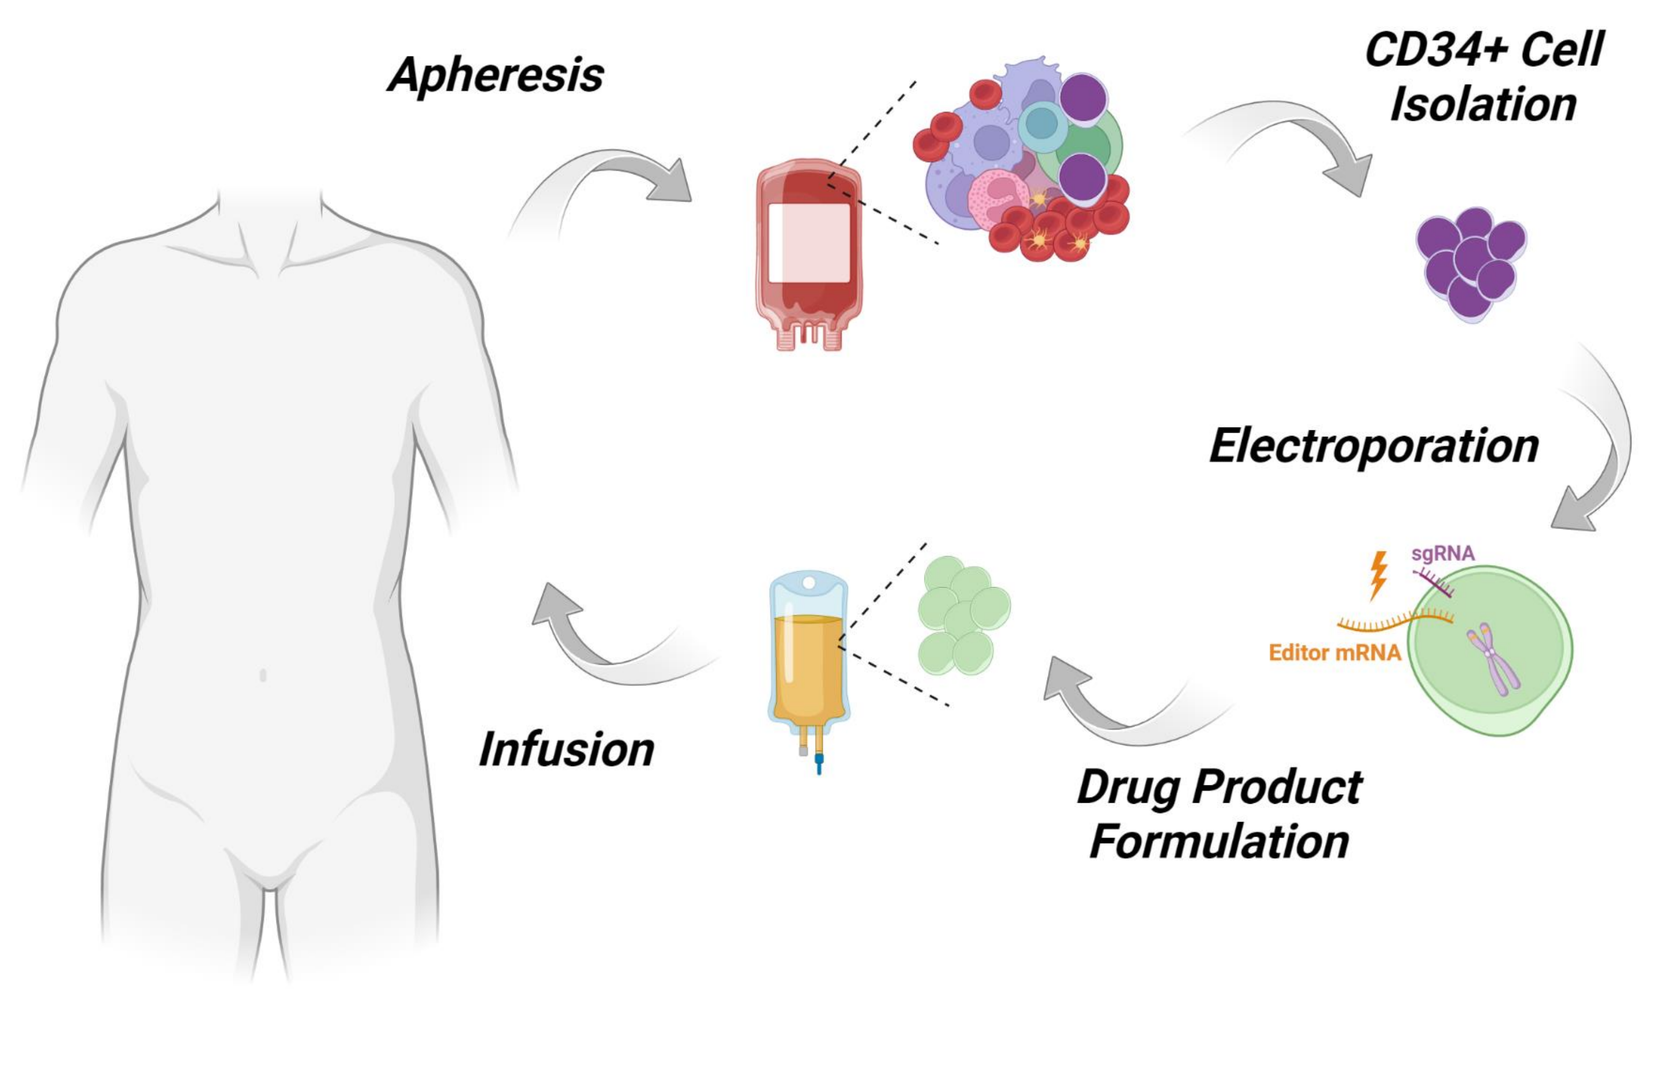
Apheresis
CD34+ Cell Isolation
Electroporation
sgRNA
Editor mRNA
Drug Product Formulation
Infusion

4. Study Methods

- ❖Nuclease-based disruption of the erythroid enhancer in the BCL11A gene was assessed at research scale. Indel formation was measured by NGS and expression of HbF was determined by high-performance liquid chromatography conducted on in vitro erythroid differentiated cells derived from ex vivo edited CD34s. Results are reported as Mean ± Standard Deviation and shown as error bars in all plots.
- ❖GMP clinical manufacturing was performed for 149 HPSC Isolations and 29 editing runs. An automated cell counter was used for cell count and viability, a flow cytometer for CD34+%, and next generation sequencing (NGS) for base editing rate. The data analysis exclude the clinical runs with known deviations. Results are reported as median, range (min – max).

5. High efficiency editing of *HBG1/2* and *BCL11A* locus

- ❖ Cas9 nuclease disruption of the *BCL11A* erythroid enhancer region (DHS+58) and *HBG1/2* promoter was assessed at research scale and compared to base editing (BE) of *HBG1/2* promoter via electroporation of mRNA of nuclease or base editor enzymes and sgRNA.
- ❖ High efficiency nuclease (>85% indels) and BE (>90% editing, <1% indels) rates were achieved in hCD34s with high viability (>95%) and high cell recovery (>90%) for all conditions.

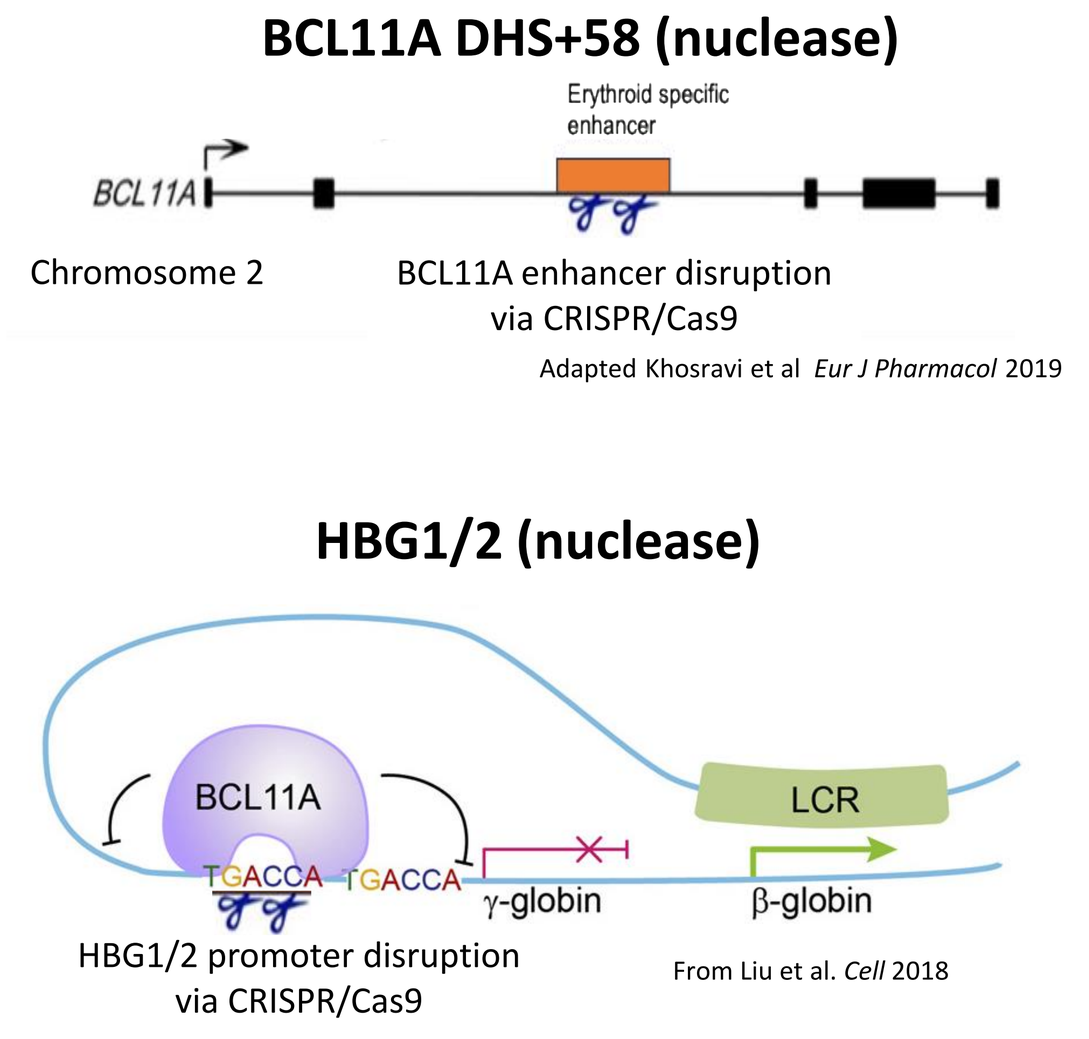
BCL11A DHS+58 (nuclease)
Erythroid specific enhancer
BCL11A
Chromosome 2
BCL11A enhancer disruption via CRISPR/Cas9
Adapted Khosravi et al Eur J Pharmacol 2019
HBG1/2 (nuclease)
BCL11A
TGACCA
LCR
γ-globin
β-globin
HBG1/2 promoter disruption via CRISPR/Cas9
From Liu et al. Cell 2018

Indel formation

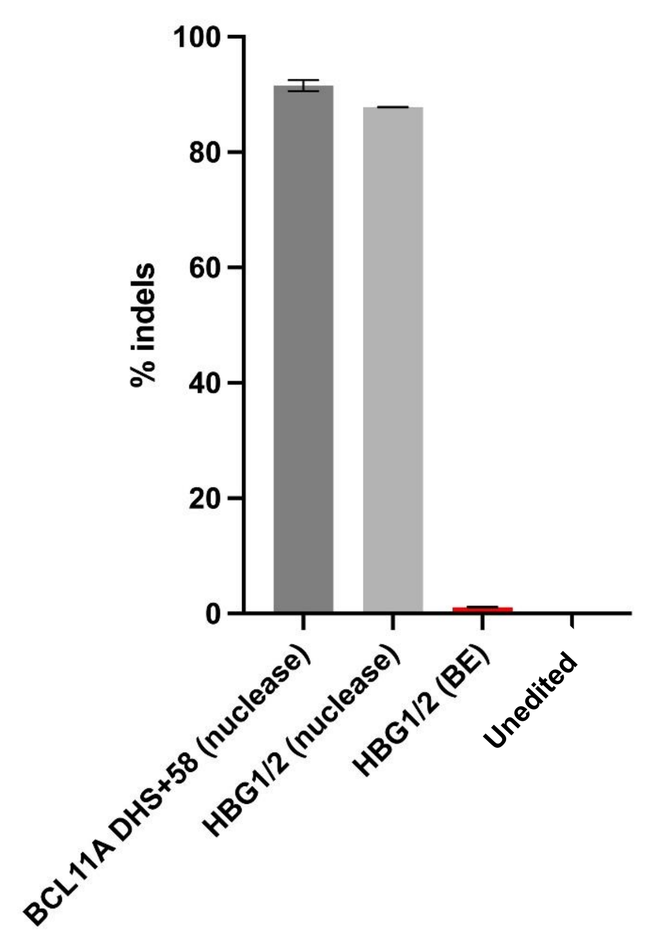
% indels
BCL11A DHS+58 (nuclease)
HBG1/2 (nuclease)
HBG1/2 (BE)
Unedited

Base editing

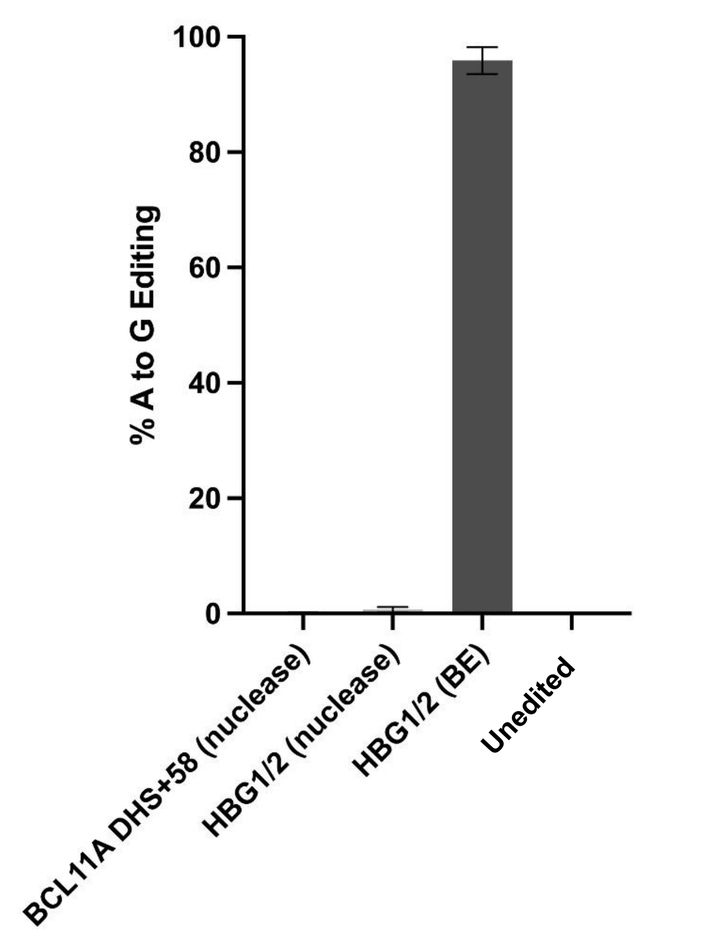
% A to G Editing
BCL11A DHS+58 (nuclease)
HBG1/2 (nuclease)
HBG1/2 (BE)
Unedited

6. Higher HbF induction with BE of HBG1/2 promoter

- ❖ BE of *HBG1/2* had 5x-fold higher induction of *HBG* expression, compared to ~2.8x fold by nuclease-based disruption of erythroid enhancer of *BCL11A* by qRT-PCR in in vitro erythroid differentiated (IVED) cells (d14)
- ❖ BE of mobilized PB hCD34s led to higher HbF induction by Ultra-High Performance Liquid Chromatography (UHPLC) for BE (60% HbF) vs nuclease-based (~40-50% HbF)

***HBB* or *HBG* expression by qRT-PCR**

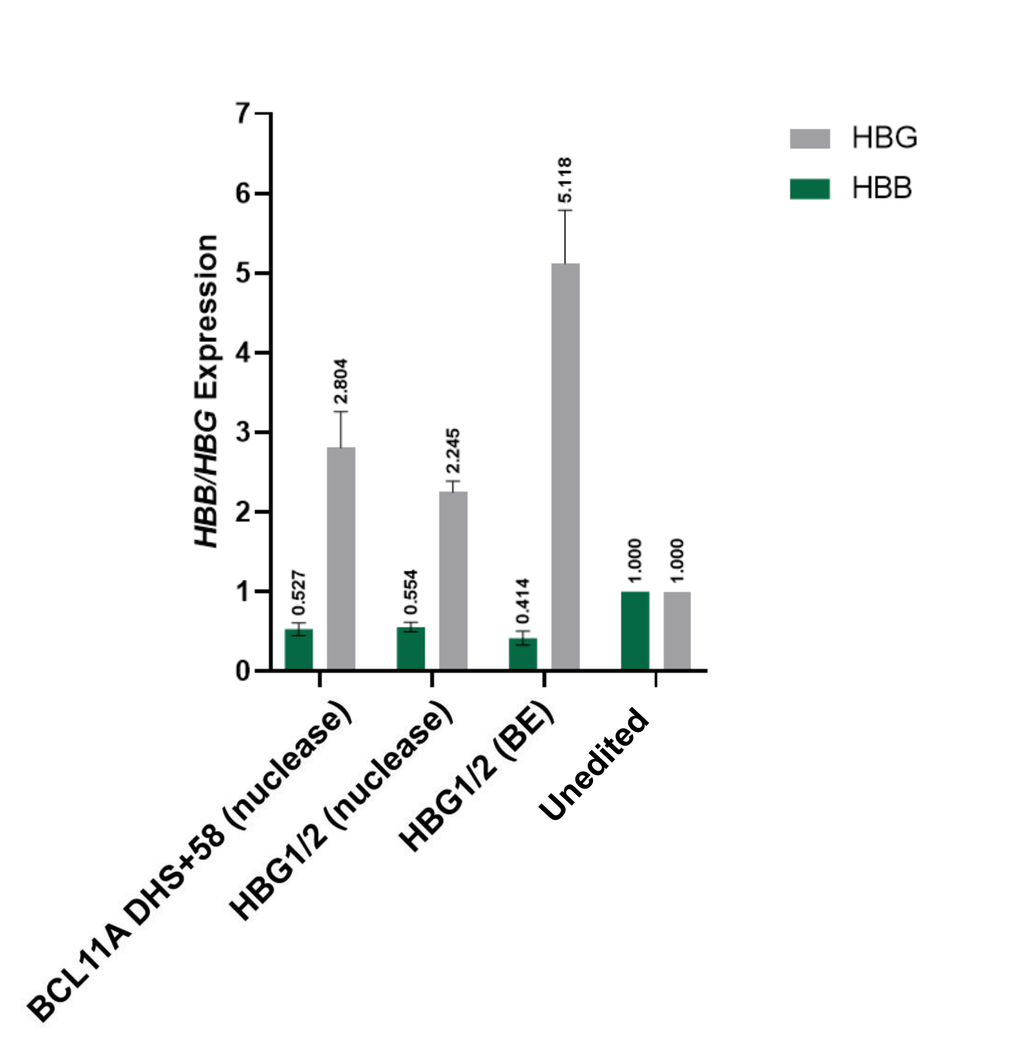
HBB/HBG Expression
HBG
HBB
0.527
2.804
0.554
2.245
0.414
5.118
1.000
1.000
BCL11A DHS+58 (nuclease)
HBG1/2 (nuclease)
HBG1/2 (BE)
Unedited

HbF Induction by UPLC

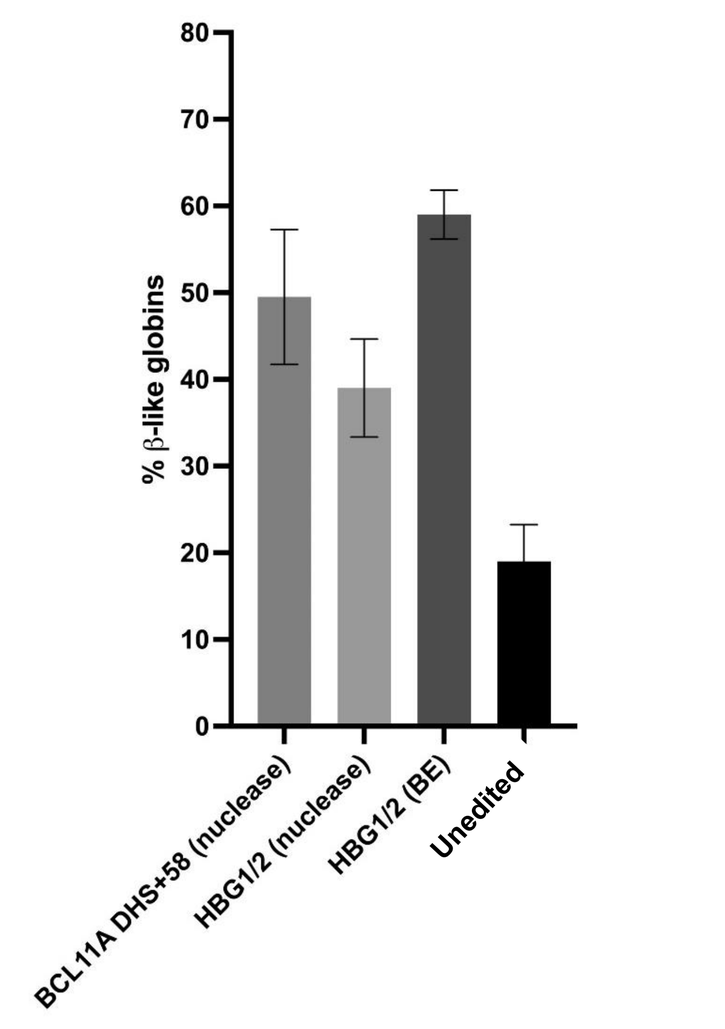
% β-like globins
BCL11A DHS+58 (nuclease)
HBG1/2 (nuclease)
HBG1/2 (BE)
Unedited

7. Nuclease erythroid enhancer disruption partially knocked down BCL11A

- ❖Nuclease based disruption (>85% indels) reduced *BCL11A* expression in IVED cells (d14) ~50% by qRT-PCR
- ❖BCL11A protein was only partially reduced despite high levels of indel formation
- ❖Lower HbF induction may be due to incomplete BCL11A knockout in erythroid cells, or possibly due to variable knockout efficiency of random insertions and deletions resulting from double strand break repair

***BCL11A* expression by qRT-PCR**

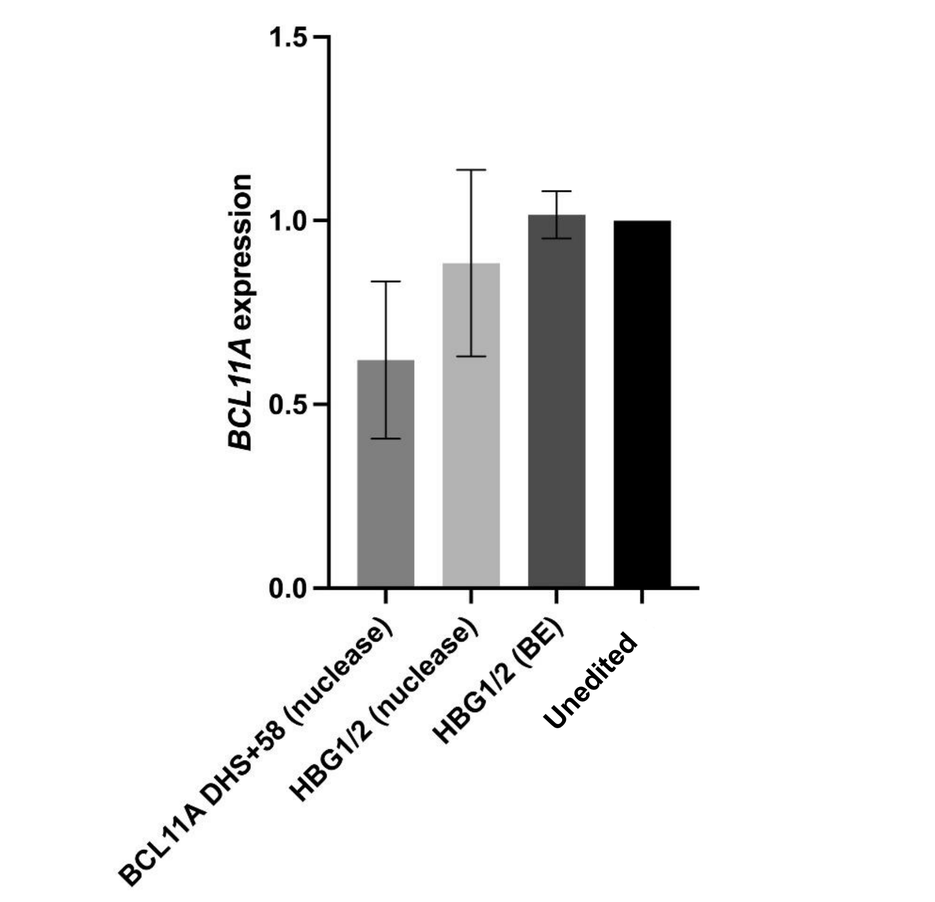
BCL11A expression
BCL11A DHS+58 (nuclease)
HBG1/2 (nuclease)
HBG1/2 (BE)
Unedited

BCL11A protein expression

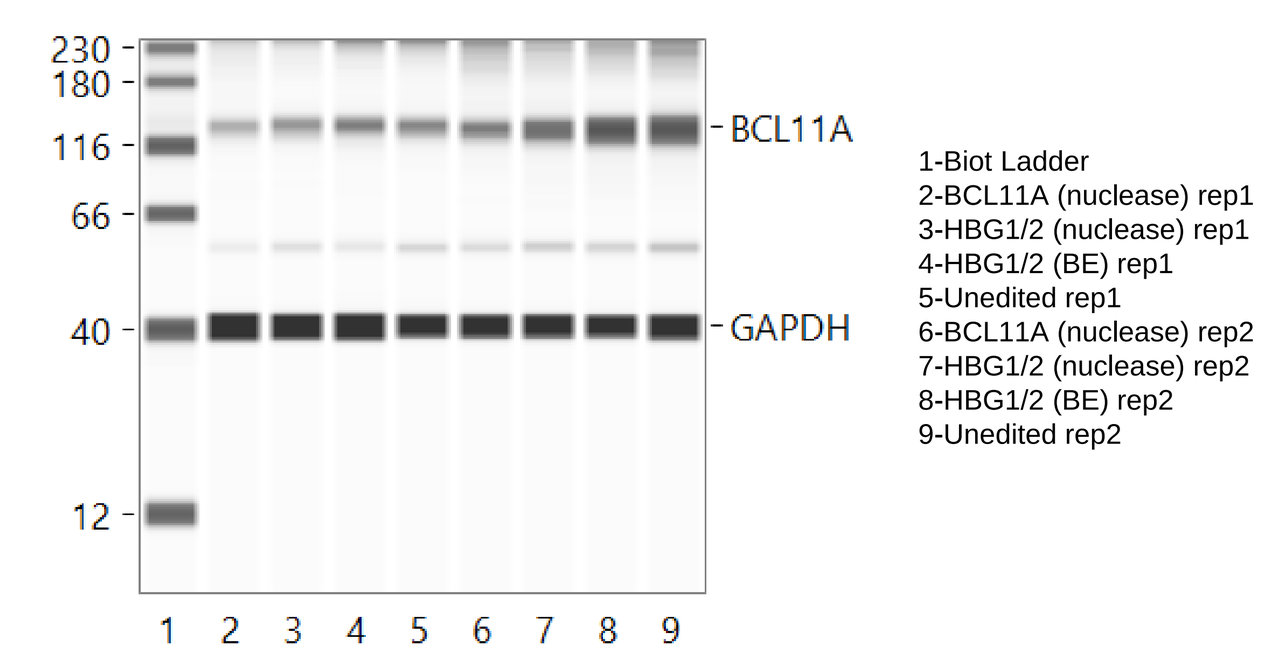
230
180
116
66
40
12
BCL11A
GAPDH
1 2 3 4 5 6 7 8 9
1-Biot Ladder
2-BCL11A (nuclease) rep1
3-HBG1/2 (nuclease) rep1
4-HBG1/2 (BE) rep1
5-Unedited rep1
6-BCL11A (nuclease) rep2
7-HBG1/2 (nuclease) rep2
8-HBG1/2 (BE) rep2
9-Unedited rep2

8. CD34+ Isolation Yield, Viability and Purity

- ❖Robust clinical isolation process performance was achieved in 149 clinical manufacturing batches with the yield (52.3%, 20.0% - 89.1%), viability (89.3%, 65.7% - 97.0%), and purity (85.6%, 52.8% - 98.6%).

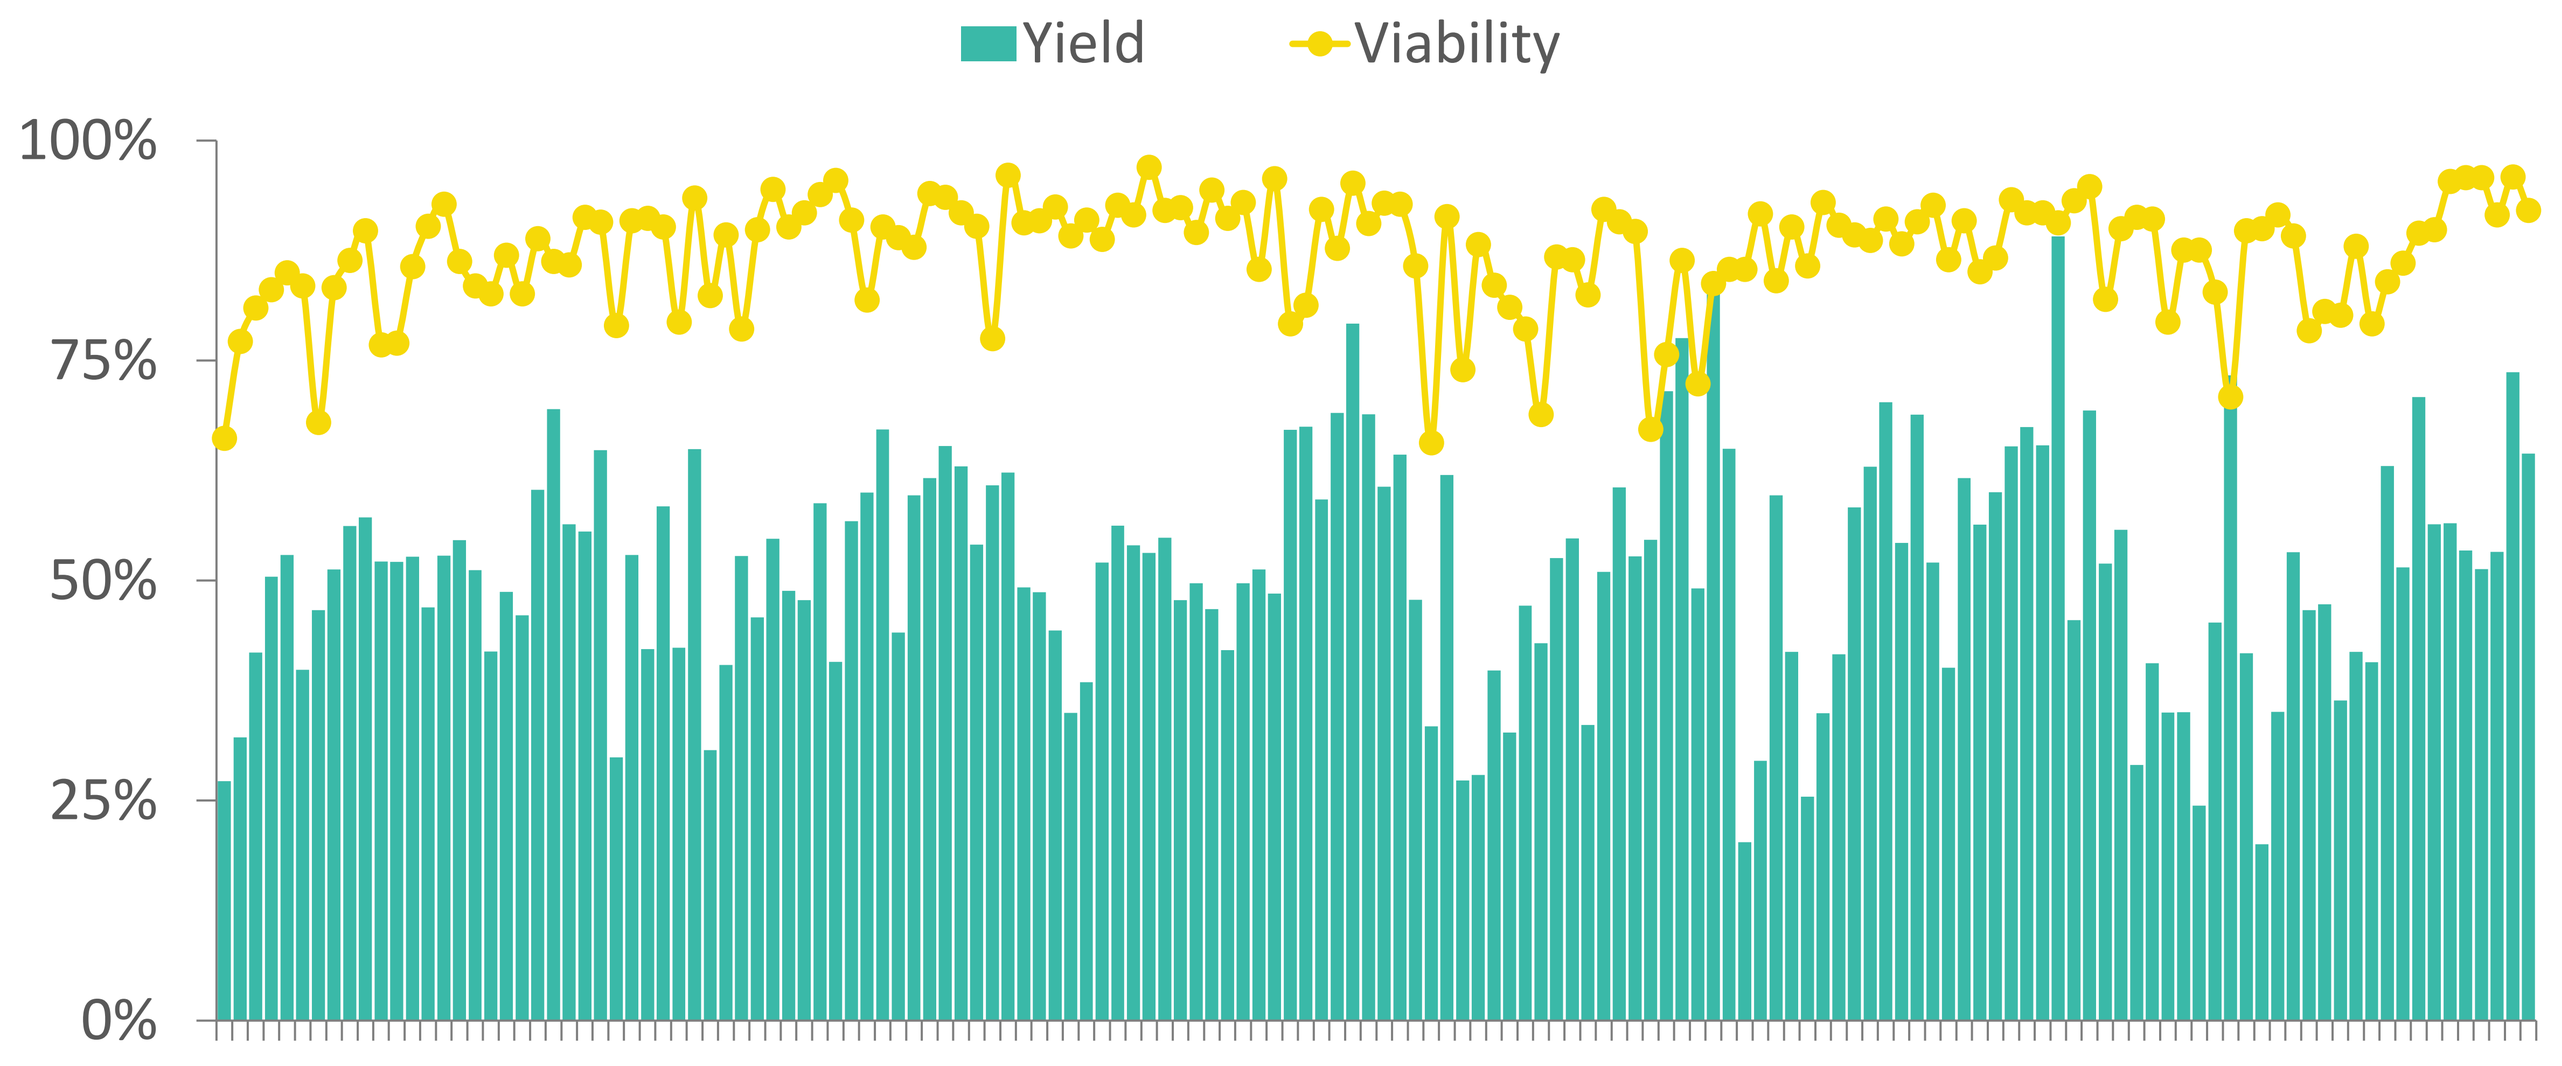
Yield
Viability
100%
75%
50%
25%
0%

9. Robust Editing Process Yield and Viability

- ❖ Consistent Editing Process yield (50.9%, 21.8% - 170.0%) and Drug Product Viability (86%, 72% - 92%) profiles were achieved in the GMP clinical runs for SCD patients.
- ❖ Editing process yield was consistent with quiescent cell culture with no evidence of multiple population doublings characteristic of differentiation, suggesting maintenance of pluripotency for BEAM-101 DP.
- ❖ BEAM-101 Dose Target was met for 100% of patients (29 of 29) on the Beacon Trial with (1, 1-3) mobilization cycles.

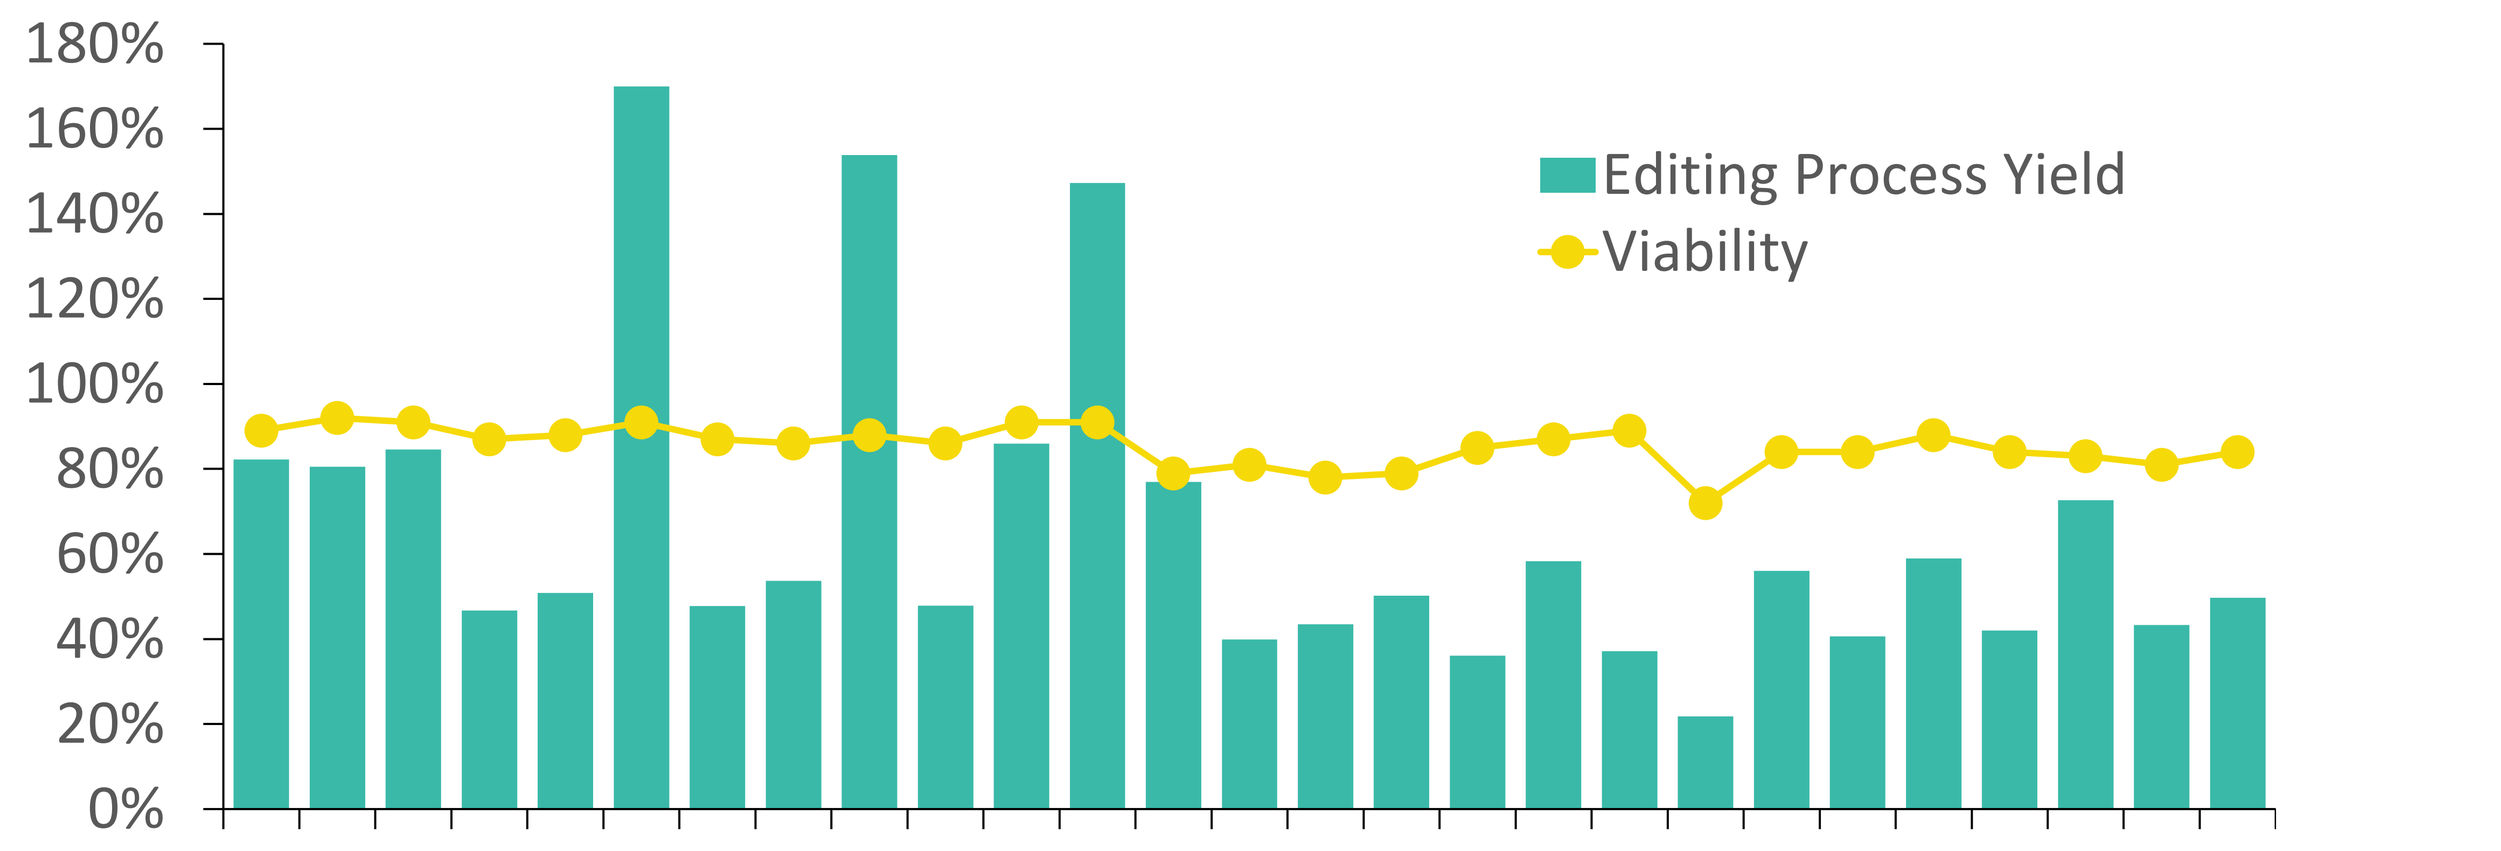
Editing Process Yield
Viability
180%
160%
140%
120%
100%
80%
60%
40%
20%
0%

10. Consistent Drug Product Quality

- ❖The BEAM-101 drug product manufactured exhibited consistently high CD34+ purity (88%, 79% - 96%) and editing rate (93%, 81% - 94%).

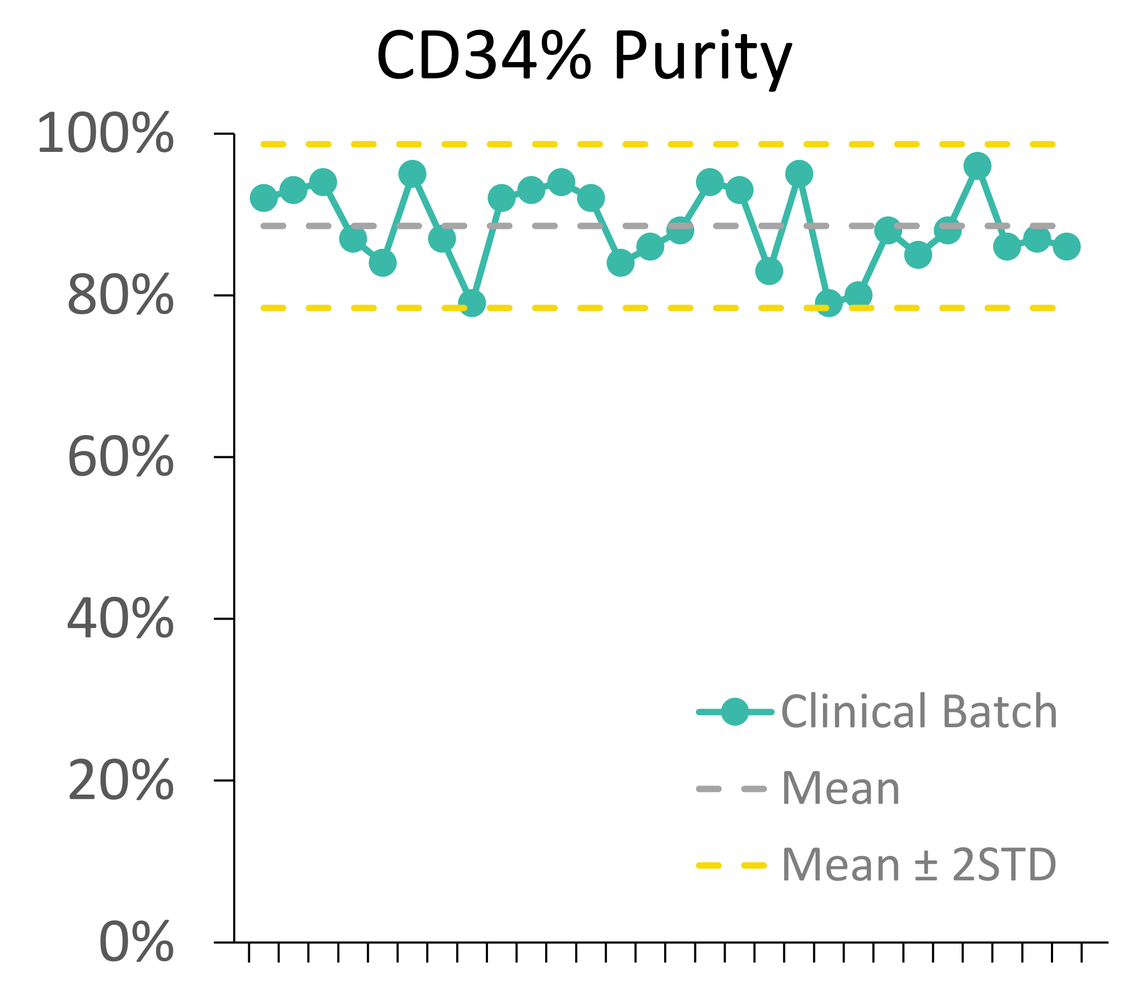
CD34% Purity
100%
80%
60%
40%
20%
0%
Clinical Batch
Mean
Mean ± 2STD

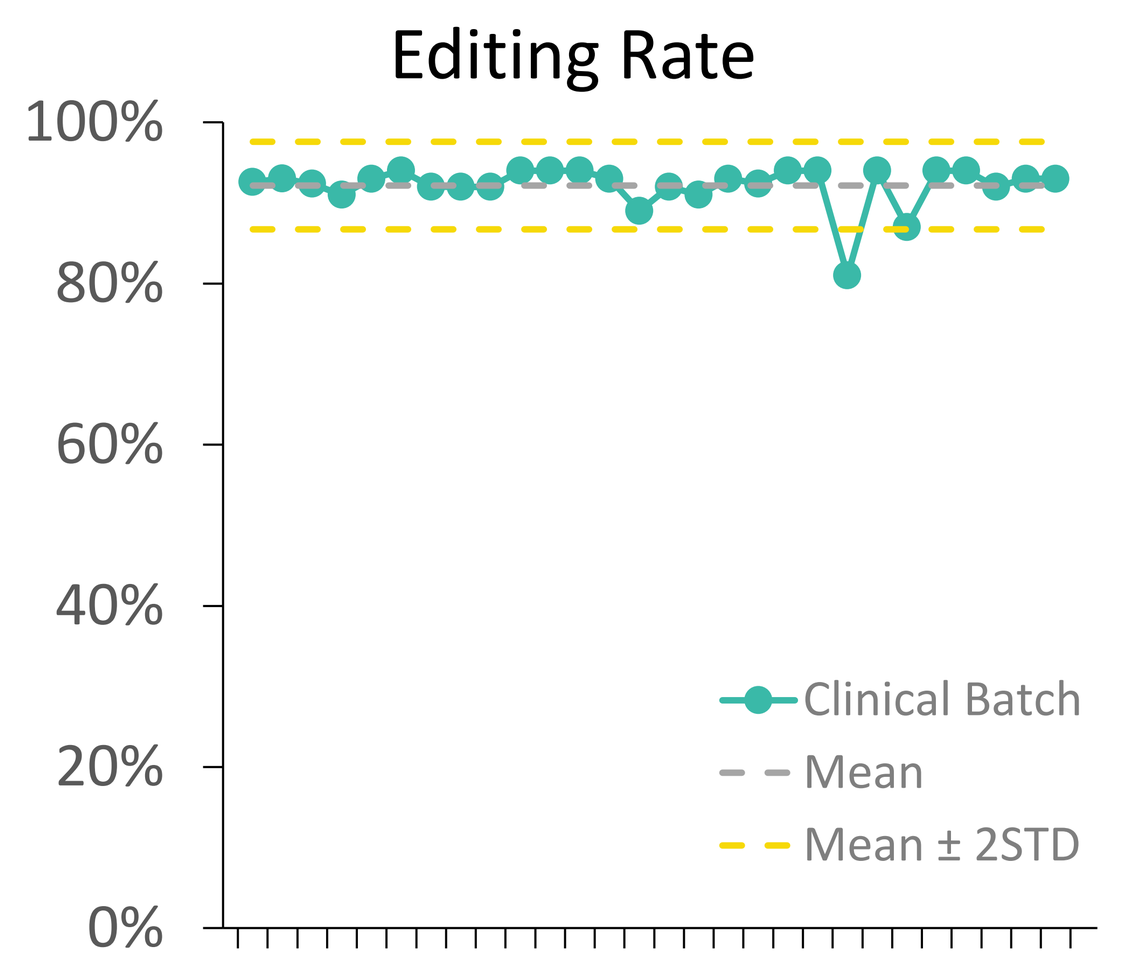
Editing Rate
100%
80%
60%
40%
20%
0%
Clinical Batch
Mean
Mean ± 2STD

11. Conclusions

- ❖BEAM-101, a CRISPR base-edited therapeutic for SCD, has been developed through the integration of process development with optimal selection of gene editing methodology.
- ❖Robust process yield, viability, and editing rates have been demonstrated while the number of mobilization cycles has been reduced.
- ❖Likewise, the selection of base editing the HBG1/2 promoter yielded superior desired fetal hemoglobin induction compared to a nuclease editing approach. The resulting process and therapeutic design delivered consistent drug product quality and potentially improved patient experience.

❖More information on BEACON clinical results for BEAM-101	**See Poster PF1151**
❖More information on BEACON biomarkers for BEAM-101	**See Poster PF1155**

12. Acknowledgement

Beam Headquarters

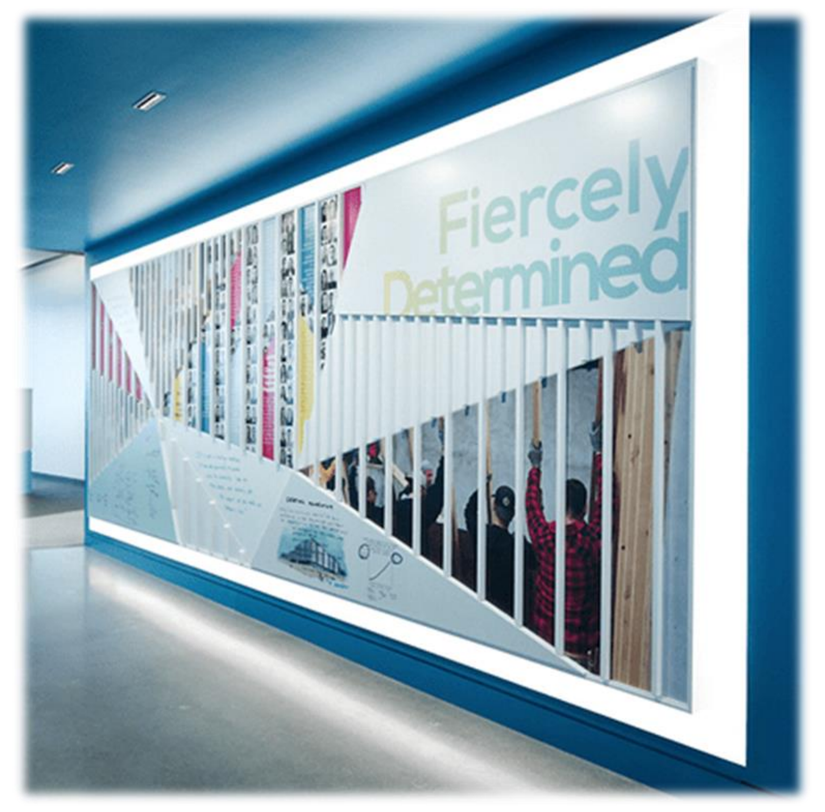
Fiercely Determined

Beam Manufacturing

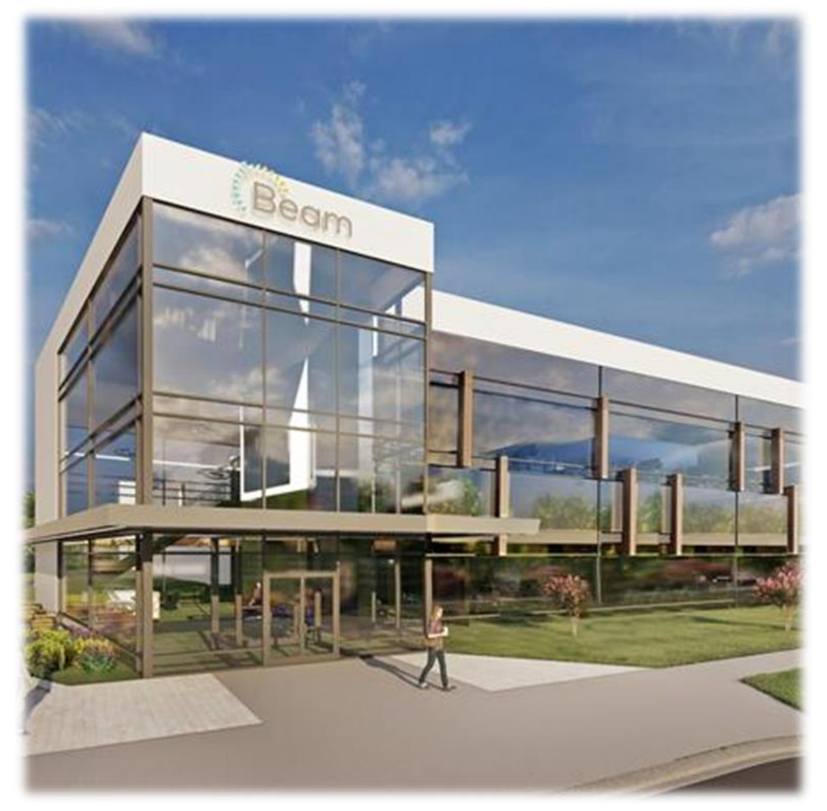
Beam

- ❖ The authors would like to thank colleagues from Beam R&D teams for their insights and discussions. We also thank colleagues from Beam Manufacturing, Quality Control, Manufacturing Science and Technology, External Manufacturing, Patient Supply, Clinical Sites, and most importantly the patients for their contributions to this work.